Poster # R6087

UNIT DOSE NASAL SPRAYS: METHOD EFFECTS OF SETTING STROKE LENGTH ON SPRAY CHARACTERISTICS

C. SHAW, M. SMITH, A. RICHIUSO, V. KULKARNI – DPT Laboratories Ltd. A. NEWCOMB – Proveris Scientific Corp.

PURPOSE

The formulation properties (viscosity, surface tension and density) and automated actuation parameters (velocity, stroke length, force and acceleration) affect the spray characteristics of unit dose nasal spray devices. During method development, various actuation mode techniques are used to determine actuation parameters. In this study, three actuation modes were compared by assessing the spray characteristics of various formulations to establish the optimal method for actuating unit dose devices.

METHODS

Unit-dose nasal spray vials (Nipro Glass Germany AG, Munnerstadt, Germany) were manually filled with six (6) different solutions: low, medium, and high viscosity glycerin and hyaluronic acid (Table 1). Manual jigs were used to insert the stoppers (West Pharmaceutical Services, Le Nouvion-en-Thierache, France) to a set depth (15.7-16.3 mm) and to assemble finished devices (vial holder and actuator: Aptar Pharma, Le Neubourg, France).

A Vereo® automated actuator (Proveris Scientific Corp., Marlborough, MA) was used to actuate the devices (Table 2) and a SprayVIEW® measurement system (Proveris Scientific) was used to measure the spray characteristics (cross-sectional area and ovality). Each of the six formulations was tested at two velocities: 50 and 70 mm/s — testing at the higher velocity was performed due to atomization concerns with the higher viscosity glycerin formulation.

THREE (3) DIFFERENT ACTUATION MODES WERE COMPARED:

1. **Position Based with Stroke Compensator:** An artificially high stroke length was set and the spring within the stroke compensator absorbed the overstroke to prevent damage to the nasal device and Vereo® actuator NSx.
2. **Position-Based Actuation With Average Stroke Length From Auto-Characterization Results:** Ten (10) automatic characterization strokes were performed on each solution using the built-in Viota® software. All sixty (60) results were averaged to determine the stroke length to be used for the subsequent testing.
3. **Force Limited Actuation:** Contact and end of stroke forces were set using a Vereo® actuator NSx operating through the Viota® Software Unit Dose Module (Proveris Scientific) with UDNSx configuration. The actuation stroke was stopped when the end of stroke force level was reached. The stroke lengths recorded are listed in Table 1.

Formulation	Viscosity at 21.75°C (cP)	Density at 22°C (g/ml)	Force Limited Actuation Stroke Length (mm): 50mm/s	Force Limited Actuation Stroke Length (mm): 70mm/s
20% Glycerin (LV Gly)	1.67	1.049	15.04 - 15.56	14.98 - 15.52
60% Glycerin (MV Gly)	10.20	1.156	15.11 - 15.43	15.22 - 15.69
70% Glycerin (HV Gly)	21.42	1.183	15.17 - 15.52	15.02 - 15.44
0.01% Hyaluronic Acid (LV HA)	3.92	1.001	15.05 - 15.47	14.43 - 15.66
0.05% Hyaluronic Acid (MV HA)	12.08	1.001	14.95 - 15.64	15.12 - 15.58
0.1% Hyaluronic Acid (HV HA)	19.99	1.001	15.00 - 15.33	14.96 - 15.58

TABLE 1

Formulation information and force-limited actuation stroke lengths

Actuation Parameter	Stroke Compensator	Automated Characterization	Force Limited Actuation
Acceleration (mm/s²)	3600	3600	3600
Velocity (mm/s)	50 / 70	50 / 70	50 / 70
Stroke Length (mm)	16	14.7	See Table 1
Actuation Force (kg)	N/A	N/A	NMT 8

TABLE 2

Actuation parameters employed

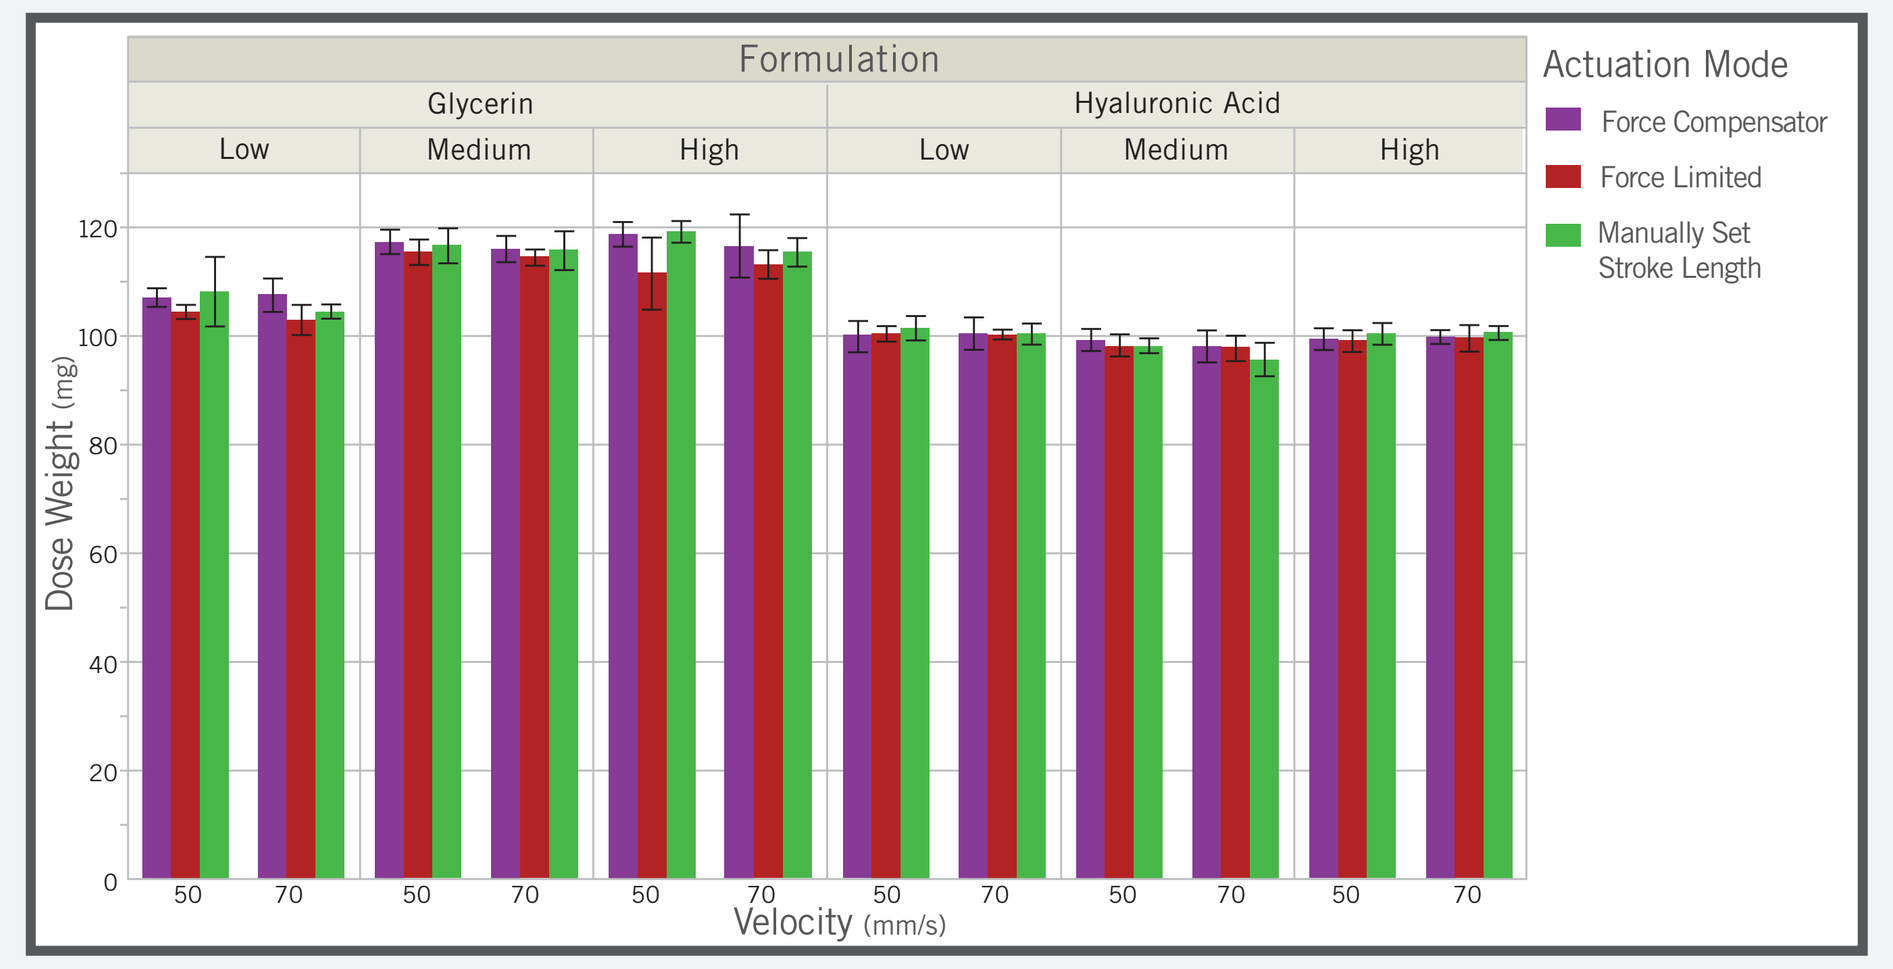
Formulation
Glycerin
Hyaluronic Acid
Low
Medium
High
Actuation Mode
Force Compensator
Force Limited
Manually Set Stroke Length
Dose Weight (mg)
Velocity (mm/s)

FIGURE 1

Comparison of actuation mode on mean dose weight for the six (6) formulations

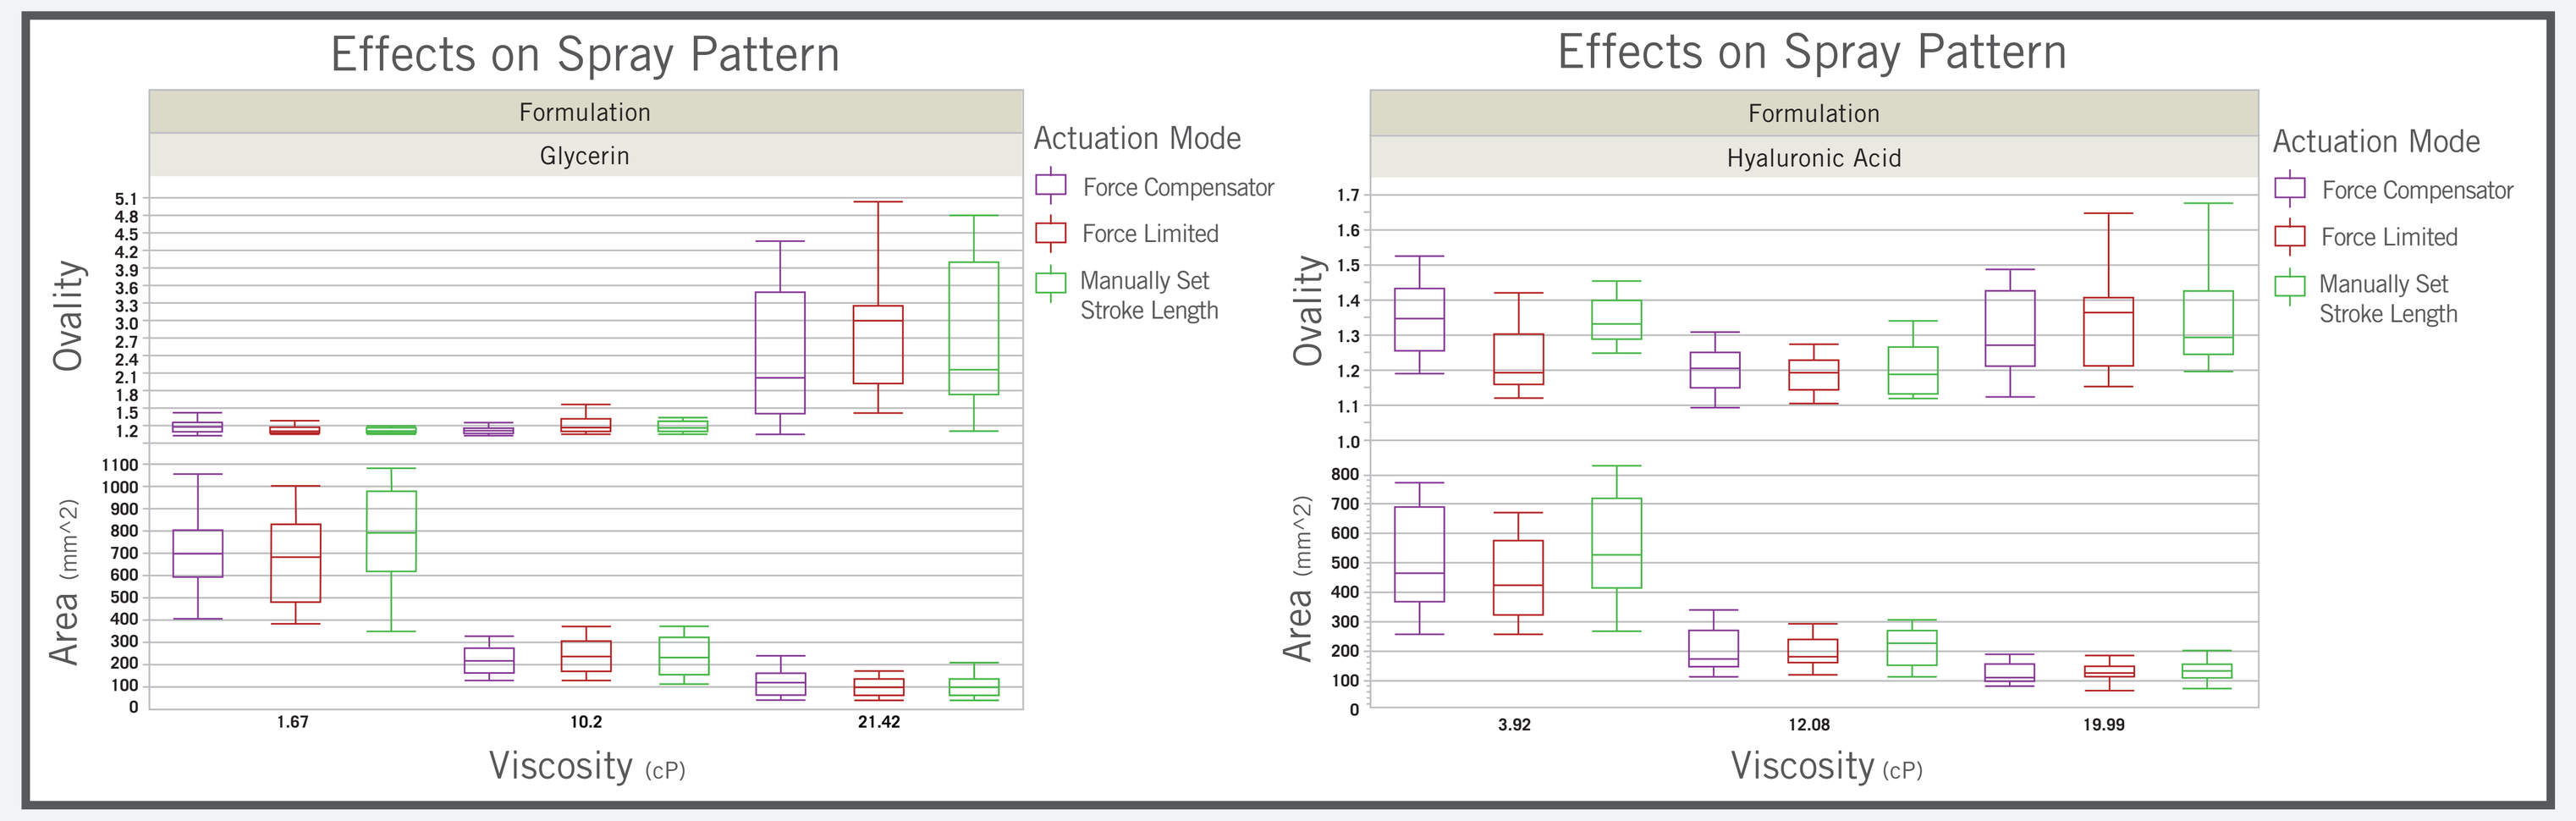
Effects on Spray Pattern
Formulation
Glycerin
Hyaluronic Acid
Actuation Mode
Force Compensator
Force Limited
Manually Set Stroke Length
Ovality
Area (mm^2)
Viscosity (cP)

FIGURE 2

Spray pattern characteristics for the six (6) formulations at both velocities

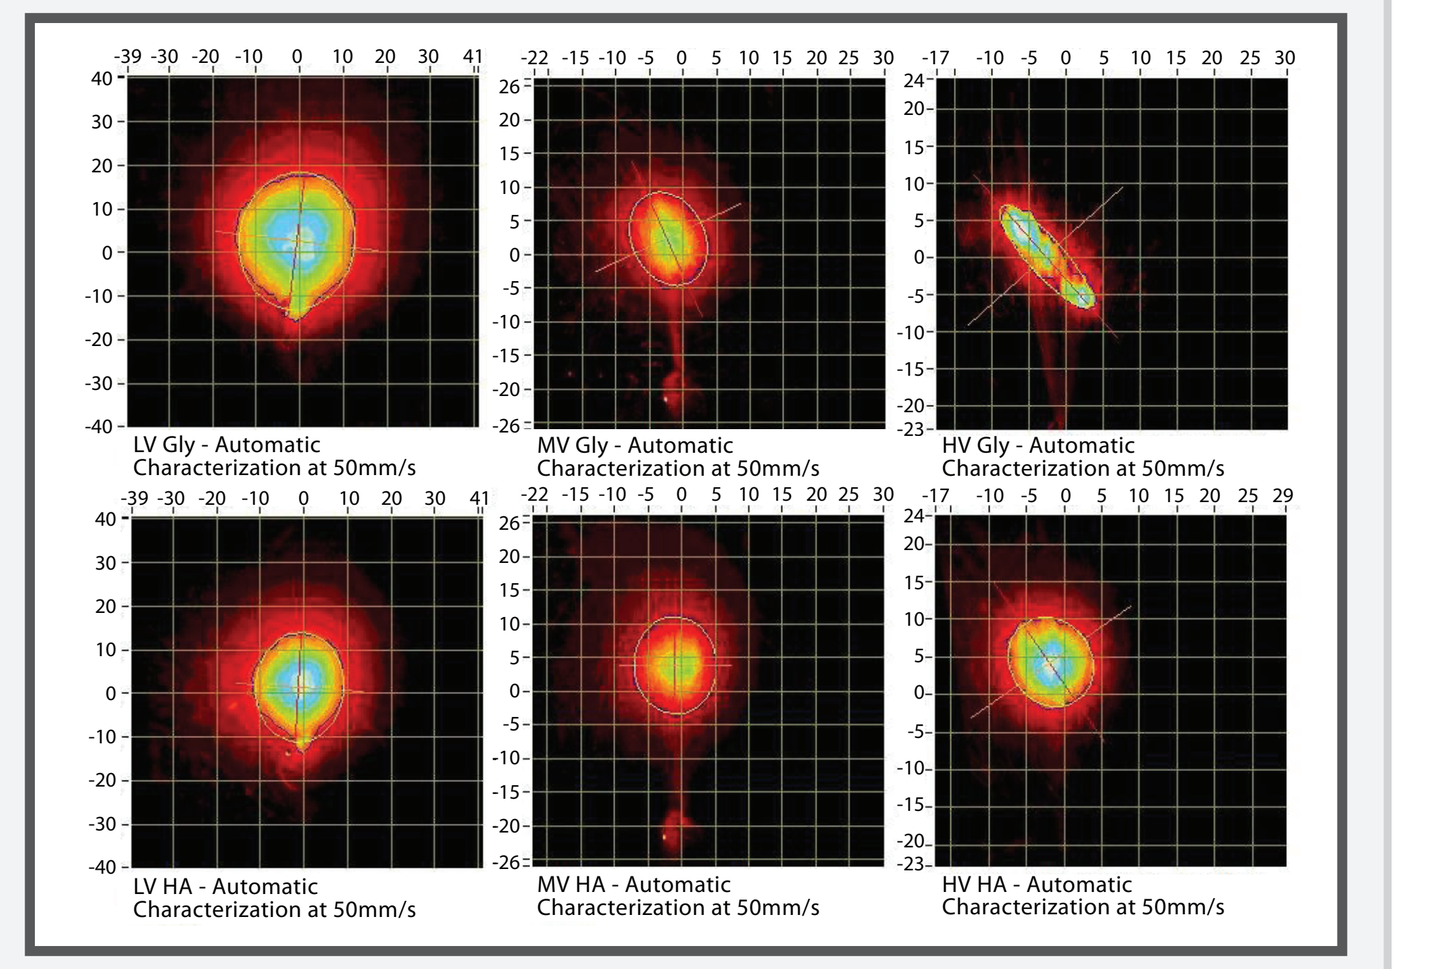
LV Gly - Automatic Characterization at 50mm/s
MV Gly - Automatic Characterization at 50mm/s
HV Gly - Automatic Characterization at 50mm/s
LV HA - Automatic Characterization at 50mm/s
MV HA - Automatic Characterization at 50mm/s
HV HA - Automatic Characterization at 50mm/s

FIGURE 3A

Examples of high-speed spray pattern images for the six (6) formulations at 50 mm/s

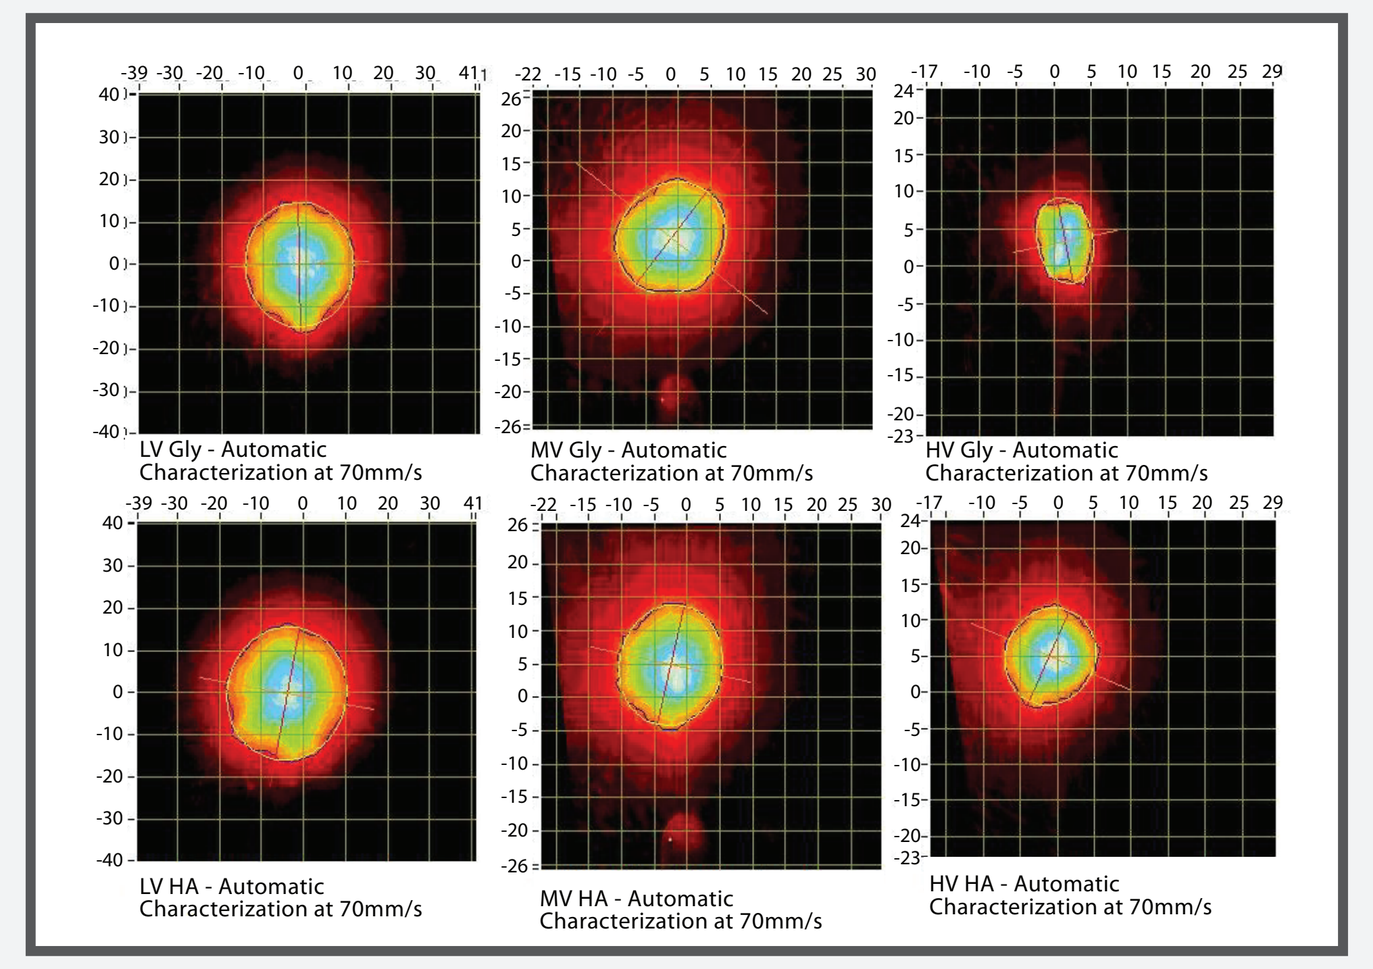
LV Gly - Automatic Characterization at 70mm/s
MV Gly - Automatic Characterization at 70mm/s
HV Gly - Automatic Characterization at 70mm/s
LV HA - Automatic Characterization at 70mm/s
MV HA - Automatic Characterization at 70mm/s
HV HA - Automatic Characterization at 70mm/s

FIGURE 3B

Examples of high-speed spray pattern images for the six (6) formulations at 70 mm/s

RESULTS

There was no significant difference observed in dose weight results in regard to the actuation mode, formulation, viscosity or actuation velocity, as shown in Figure 1. This shows that all three tested actuation modes were capable of actuating the devices successfully to achieve the desired dose weight. However, from the spray pattern cross-sectional area and ovality results in Figure 2, it becomes clear that these parameters are dependent on the viscosity of the formulation. When using a unit dose device with a viscous formulation, the actuation results in a "squirt-type" manner where the cross-sectional area is smaller with larger ovality. Figures 3A and 3B show examples of high-speed images of the spray pattern for the different formulations at both actuation velocities.

CONCLUSIONS

- Dose weight appears to be relatively independent of the actuation methods evaluated and the formulation viscosities. As long as the stopper is fully depressed into the vial, dose weight will be a function of the vial fill weight.
- The cross-sectional area and ovality of the spray pattern are highly dependent on formulation viscosity.
- Increasing the actuation velocity reduced the ovality and increased spray area.
- Proveris Viota® Software Unit Dose Module proved to be the most beneficial actuation mode. It prevented both under- and over-actuating the unit dose device, and eliminated the need to waste filled devices to find an average stroke length for testing.

REFERENCES

- Dayal P., Shaik M.S., and Singh M. "Evaluation of different parameters that affect droplet size distribution from nasal sprays using the Malvern Spraytec" J. Pharm. Sci. 93: 1725-1742 (2004)
- Kulkarni V.S., Shaw C., Smith M., and Brunotte J. "Characterization of plumes of nasal spray formulations containing mucoadhesive agents sprayed from different types of devices" Poster at AAPS Annual Meeting (2013)
- Trows S.,Wuchner K.,Spycher R., and Steckel H. "Analytical Challenges and Regulatory Requirements for Nasal Drug Products in Europe and the U.S." Pharmaceutics. 6(2): 195–219 (2014)